Development and pre-clinical efficacy characterization of a systemically administered multiple Toll-like receptor (TLR) agonist for anti-tumor immunotherapy

DO NOT POST

DO NOT POST

Michael J. Newman, Ph.D.

Decoy Biosystems, San Diego, CA (Resident of JLABS)

Abstract/Poster #B178

CRI-CIMT-EATI-AACR 2018

Background and Methods

BACKGROUND) The first cancer immunotherapy was developed in the 1890's by Dr. William Coley, who observed tumor regressions after injecting cancer patients with a heat-killed mixture of Gram-positive and negative pathogenic bacteria. Dr. Coley determined that the Gram-negative bacteria were the principal contributor to anti-tumor activity and that the product would likely be most effective when administered intravenously (i.v.), but was too toxic in this setting, limiting use to intramuscular and intratumoral administration. Coley's toxins, as it was known, was credited with curing hundreds of late stage cancer patients over 70 years. However, lack of knowledge regarding mechanism of action made it difficult to optimize or standardize, producing high variability. The use of multiple, local administration routes also likely contributed to variability in response, leading the FDA to refuse to grandfather in the product in 1963. We now know the mechanism of action of Coley's toxins and the source of the i.v. toxicity. Gram-negative bacteria contain immune system danger signals, including multiple TLR agonists (activating TLRs 2, 4, 5 and 9), which directly and indirectly, via induction of cytokine and chemokine secretion, participate in the activation of most of the cellular mediators of innate and adaptive immune responses. Lipopolysaccharide (LPS), which activates TLR4, has been identified as a major contributor to both the anti-tumor activity and i.v. toxicity of Gram-negative bacteria. Decoy's hypothesis is that significant reduction without complete elimination of LPS activity, in conjunction with killing and stabilization of non-pathogenic, Gram-negative bacteria may produce a multiple TLR product that can safely and effectively induce anti-tumor immune responses via i.v. administration.

MATERIALS AND METHODS) Product manufacturing (Decoy10) and MTD in mice were carried out by Molecular Diagnostic Services (San Diego, CA). Non-pathogenic, diaminopimelic acid (DAP) auxotrophic *E. coli* strain 2617-143-312 (Migula) Castellani and Chalmers (ATCC 13070) was grown in LB/Miller broth supplemented with 0.5% glucose, 1 mM DAP and 2 mM MgCl2. Late log phase cells were harvested, washed twice with LB/Miller broth, 0.1 mM DAP, 20 mM MgCl2 at 4^oC by centrifugation and resuspended at 1x10^10 cells per ml. LPS activity was reduced by treating cells with 1 mg/ml polymyxin B (Calbiochem #5291) for 1 hour at 4^oC, with gentle stirring. Cells were washed three times with 4^oC PBS, pH 7.5, 20 mM MgCl2, resuspended at 1x10^10 per ml, and additional pyrogens were inactivated, and the cells were killed, by incubation with 1% glutaraldehyde (GA) for 1 hour with gentle stirring at 4^oC. Cells were washed three times in above incubation medium without GA, resuspended in 50% PBS, pH 7.5, 2 mM MgCl2, 12% trehalose (freezing medium), flash-frozen and stored at -80^oC. Viability and strain confirmation were assessed by plating efficiency $\pm$DAP. LPS/endotoxin activity was determined by Limulus Amebocyte Lysate (LAL) assay. Cellular integrity was assessed by electron microscopy (EM) and/or optical microscopy after Gram staining. Pyrogenicity was assessed in a standard rabbit rectal temperature test by Pacific BioLabs, Inc. (Hercules, CA).

In vivo anti-tumor assays were carried out by Southern Research Institute (Birmingham, AL), Crown Bioscience (San Diego, CA and Beijing, China) and AntiCancer, Inc., (San Diego, CA) with ~7 week old female BALB/c, C57BL/6 or CB17/SCID mice. Decoy10 was washed by centrifugation and resuspended in PBS, 2 mM MgCl2 prior to *in vivo* administration. Anti-tumor activity was assessed in orthotopic CT26 murine colorectal carcinoma (tumor fragments sewn onto secum), subcutaneous (s.c.) A20 murine non-Hodgkin's lymphoma (NHL), s.c. human Ramos NHL, and s.c. H22 murine hepatocellular carcinoma (HCC).

RESULTS) Treatment of non-pathogenic, Gram-negative *E. coli* with polymyxin B and glutaraldehyde under conditions to kill and stabilize the cells (Decoy10) resulted in >90% reduction of *in vitro* LPS/endotoxin activity and *in vivo* pyrogenicity. Decoy10 also exhibited a 3-fold reduction in acute *in vivo* toxicity relative to untreated bacteria. Surprisingly, induction of anti-tumor cytokine secretion by human peripheral blood mononuclear cells (PBMCs) was not compromised, relative to untreated bacteria. Treatment with Decoy bacteria i.v. produced significant single agent anti-tumor activity against orthotopic murine colorectal carcinoma and metastatic murine pancreatic carcinoma. Synergistic combination activity, including eradication of established tumors, with a therapeutic index of up to 10-fold, was observed in combination with IL-2 or low-dose cyclophosphamide (LDC) in murine colorectal carcinoma models, with LDC in an s.c. murine non-Hodgkin's lymphoma (NHL) model and with LDC plus rituximab in an s.c. human NHL model. Synergistic anti-tumor activity was also observed in combination with a low-dose, non-steroidal anti-inflammatory drug (NSAID) in a metastatic, murine pancreatic carcinoma model (data not shown). In addition, tumor eradications were observed in combination with NSAID and were enhanced by addition of anti-PD1 therapy in an s.c. murine hepatocellular carcinoma model. Optimal (80-100%) tumor eradication was shown to be mediated by natural killer (NK), CD4+ and CD8+ T cells. Immunological memory (80-100% and partial), determined by rejection of subsequent tumor challenge, was demonstrated in both immune competent and innate only settings, respectively.

Table 1. Decoy treatment kills *E. coli* and significantly reduces LPS endotoxin activity, pyrogenicity and acute i.v. toxicity

Treatment	Live Bacteria	LPS/Endotoxin Activity LAL Assay	Pyrogenicity Threshold Rabbit Assay
No Treatment	82.9%	44.7 Units / 10^6 Bacteria	3 x 10^4 Bacteria
Decoy10	0	3.6 Units / 10^6 Bacteria	9 x 10^5 Bacteria
Change induced by treatment	Killed all bacteria	12-fold or 92% reduction	30-fold or 97% reduction

i.v. MTD in mice of untreated bacteria 3x10^9 / Decoy10 1x10^10 (data not shown)

Results

Figure 1. Decoy-treated bacteria are intact and relatively monodisperse

Untreated Bacteria | Decoy-Treated Bacteria

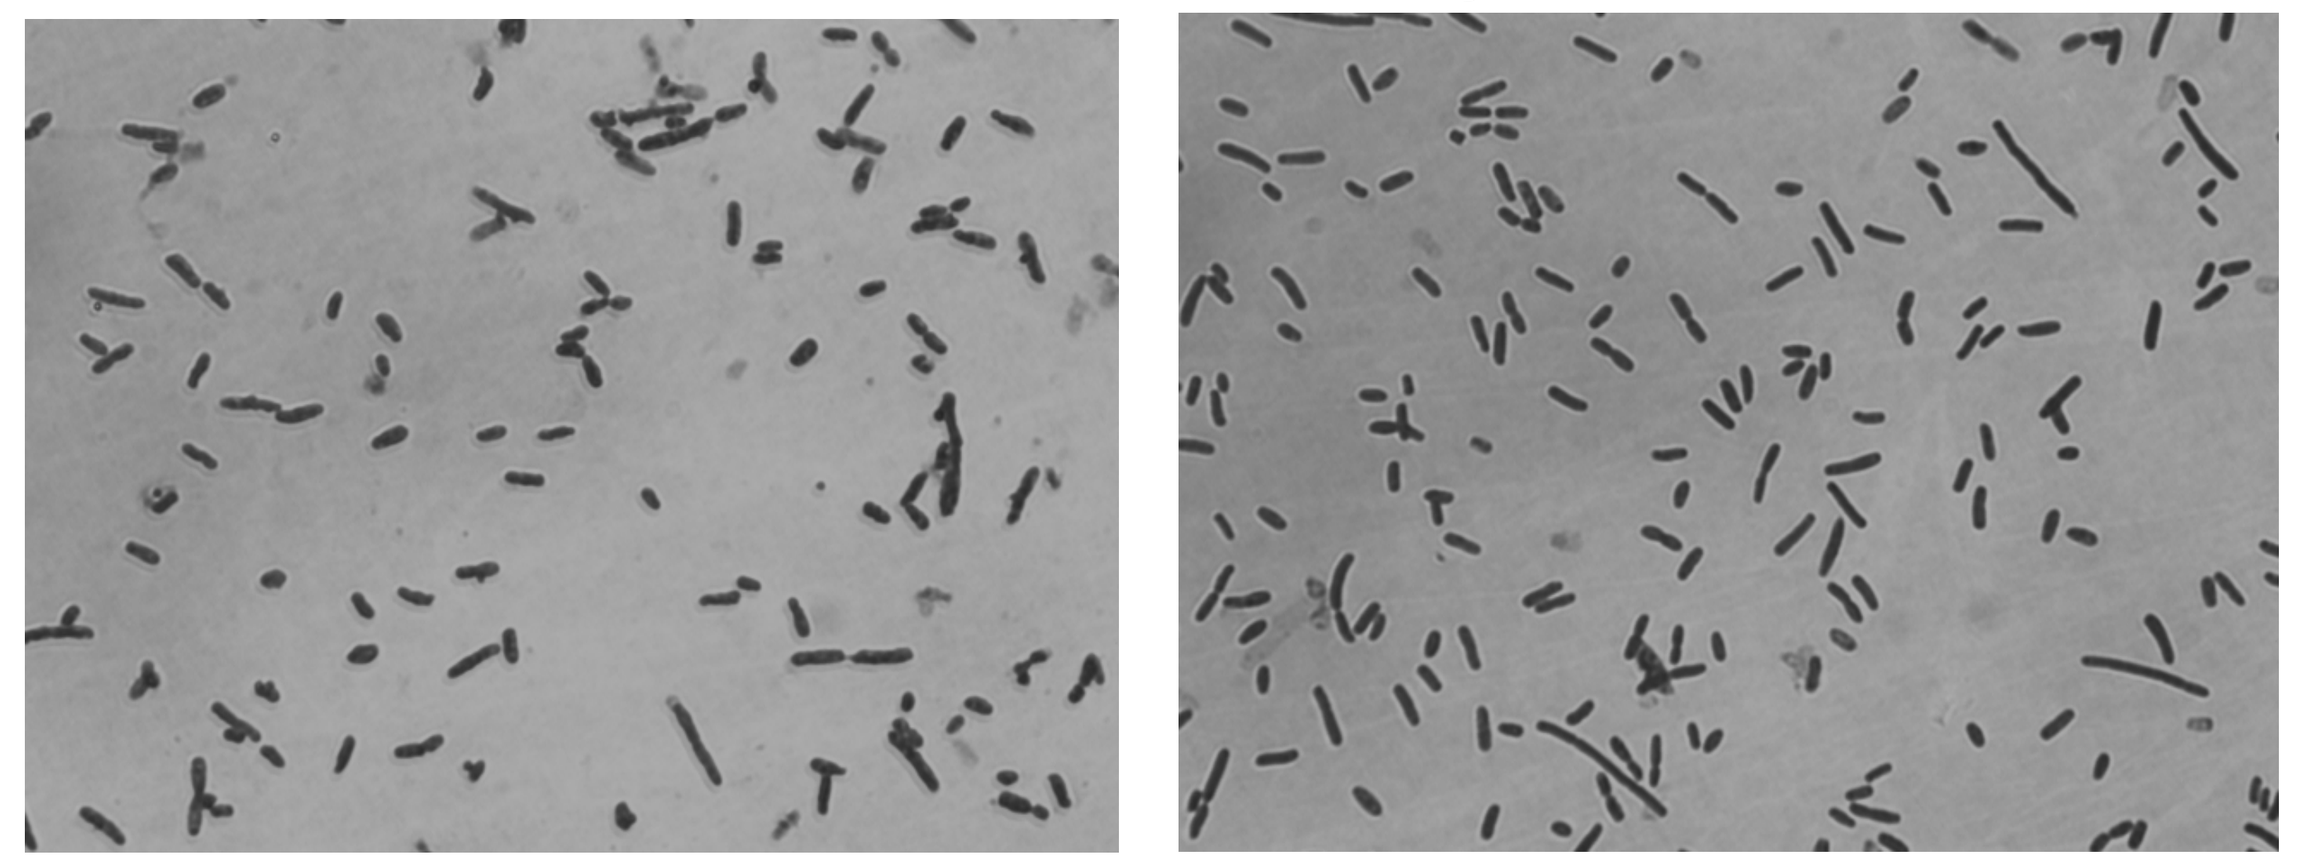

Gram-Stained Bacteria / Light Microscope Photographs

Table 2. Decoy10 induces human immune cells to secrete higher levels of most anti-tumor cytokines than untreated bacteria

Secretion by Human PBMCs *In Vitro*	Untreated Bacteria	Decoy-Treated Bacteria (Decoy10)
Anti-Tumor Cytokine	48 hour pg/mL peak (mean of triplicates) at same bacteria dose for each cytokine	
GM-CSF	1,094	1,197
IFNγ	175,866	47,488*
IL-12p70	176	528
IL-23	0	119
TNFα	49,782	77,919

PBMCs from fresh normal peripheral blood leukapheresis paks (ALLCells, Alameda, CA) were isolated using a Ficoll gradient. Ten-fold increments of 1x10/mL to 1x10^8/mL untreated or Decoy10 bacteria were incubated with 2.5x10^5 PBMCs for 48 hours. Luminex analysis of supernatants was carried out using Millipore human cytokine/chemokine magnetic bead panels. *Results presented are the peak levels determined for each cytokine, which occurred at the same untreated and treated bacterial dose, except for INFγ, which peaked at a lower dose for the untreated bacteria and is compared to the same dose of Decoy-treated bacteria. Decoy-treated bacteria (Decoy10) contained 4.94% as much LPS/endotoxin as the untreated bacteria on a per bacterium basis as determined by the LAL assay. The experiment was carried out by Eurofins Panlabs, Inc. (St. Charles, MO).

Table 3. Decoy10 induces human immune cells to secrete higher levels of anti-tumor cytokines than mono-specific TLR agonists

	CpG ODN (TLR9a)	Poly(I:C) (TLR3a)	R848 (TLR 7/8a)	LPS (TLR4a)	Decoy10
Anti-Tumor Cytokine	pg/mL (48 hour full titration peak mean)				
GM-CSF	0	0	87	175	1,197
IFNγ	7	103	31,324	29,416	91,475
IL-12p70	4	18	253	109	528
TNFα	51	208	33,393	24,944	77,919

The experiment was carried out by Eurofins Panlabs as described for Table 2. Toll-like receptor agonists (TLRa) were obtained from InvivoGen (San Diego, CA) and titrated in the experiment as follows; CpG ODN 2006 (#tlrl-2006, 0.005 to 5 micromolar), Poly(I:C) (#tlrl-pic, 0.001 to 100 microgram/mL), R848 (#tlrl-r848, 0.1 to 100 microgram/mL) and *E. coli* LPS (#tlrl-pb5lps, 10 to 1x10^6 picogram/mL). TLR agonist stocks were prepared as recommended by the manufacturer at up to their recommended limits of solubility and results are the peak cytokine levels determined for each TLRa and Decoy10.

Michael J. Newman / 1-858-922-9015 / mnewman@decoybio.com

Results

Figure 2. Single agent Decoy10 inhibits metastasis and extends survival of mice with orthotopic CT26 mouse colorectal carcinoma

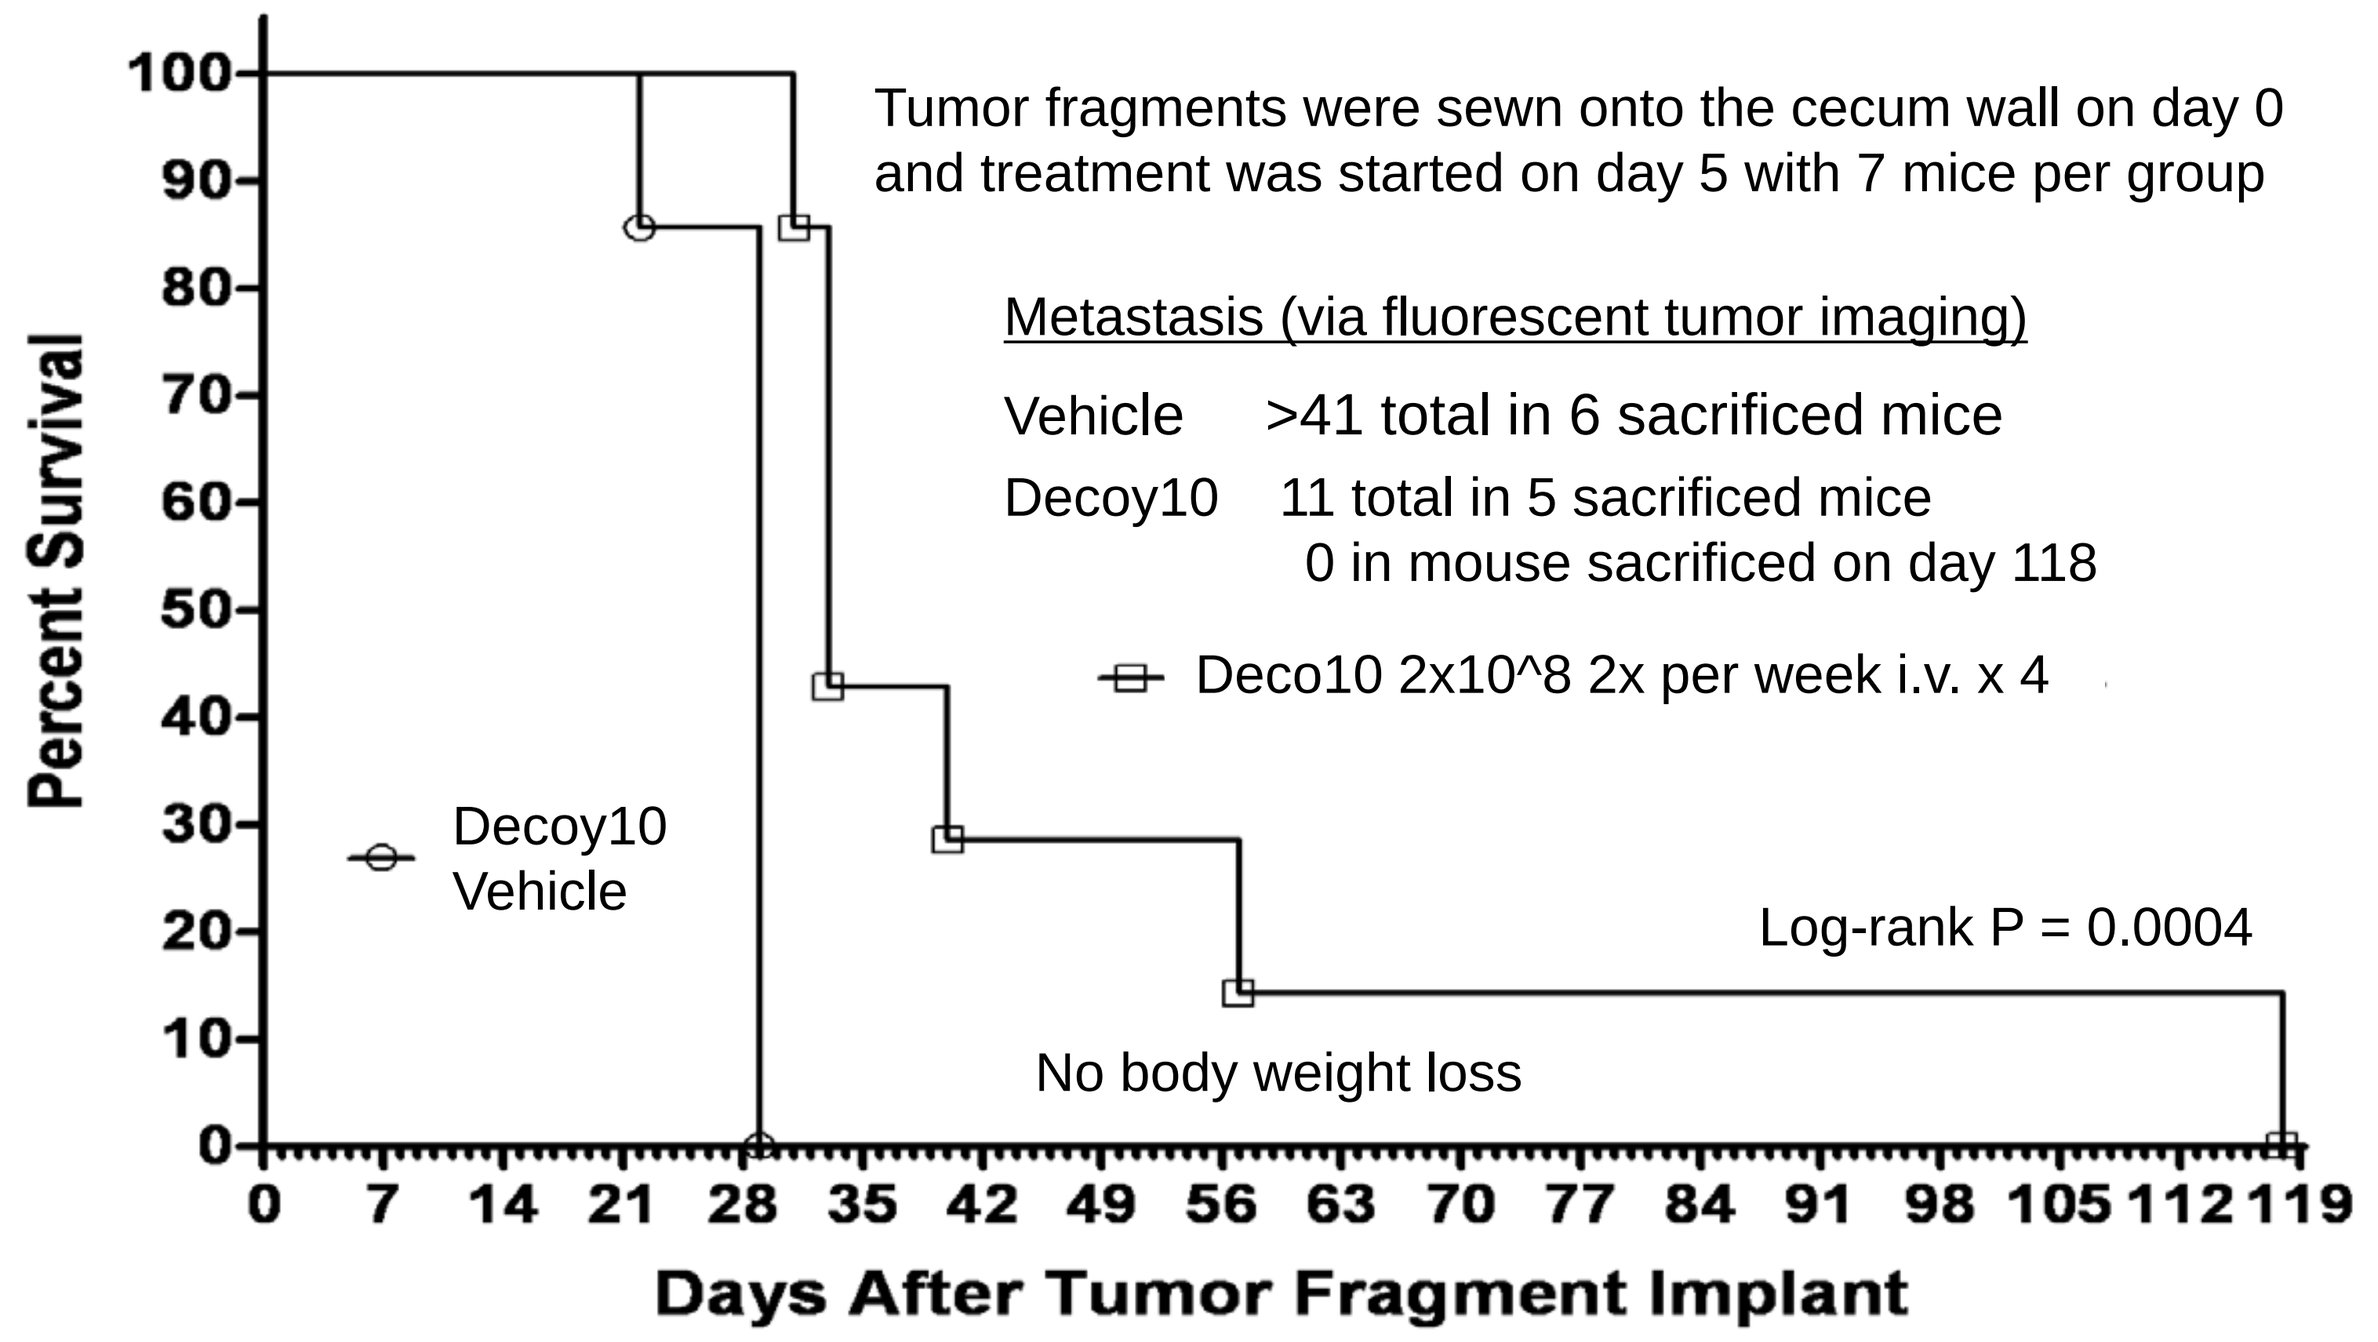
Percent Survival
Tumor fragments were sewn onto the cecum wall on day 0 and treatment was started on day 5 with 7 mice per group
Metastasis (via fluorescent tumor imaging)
Vehicle >41 total in 6 sacrificed mice
Decoy10 11 total in 5 sacrificed mice
0 in mouse sacrificed on day 118
Deco10 2x10^8 2x per week i.v. x 4
Decoy10
Vehicle
Log-rank P = 0.0004
No body weight loss
Days After Tumor Fragment Implant

Figure 3. Decoy10 synergizes with low-dose cyclophosphamide (LDC) to eradicate 200 mm³ established s.c. A20 mouse NHL tumors

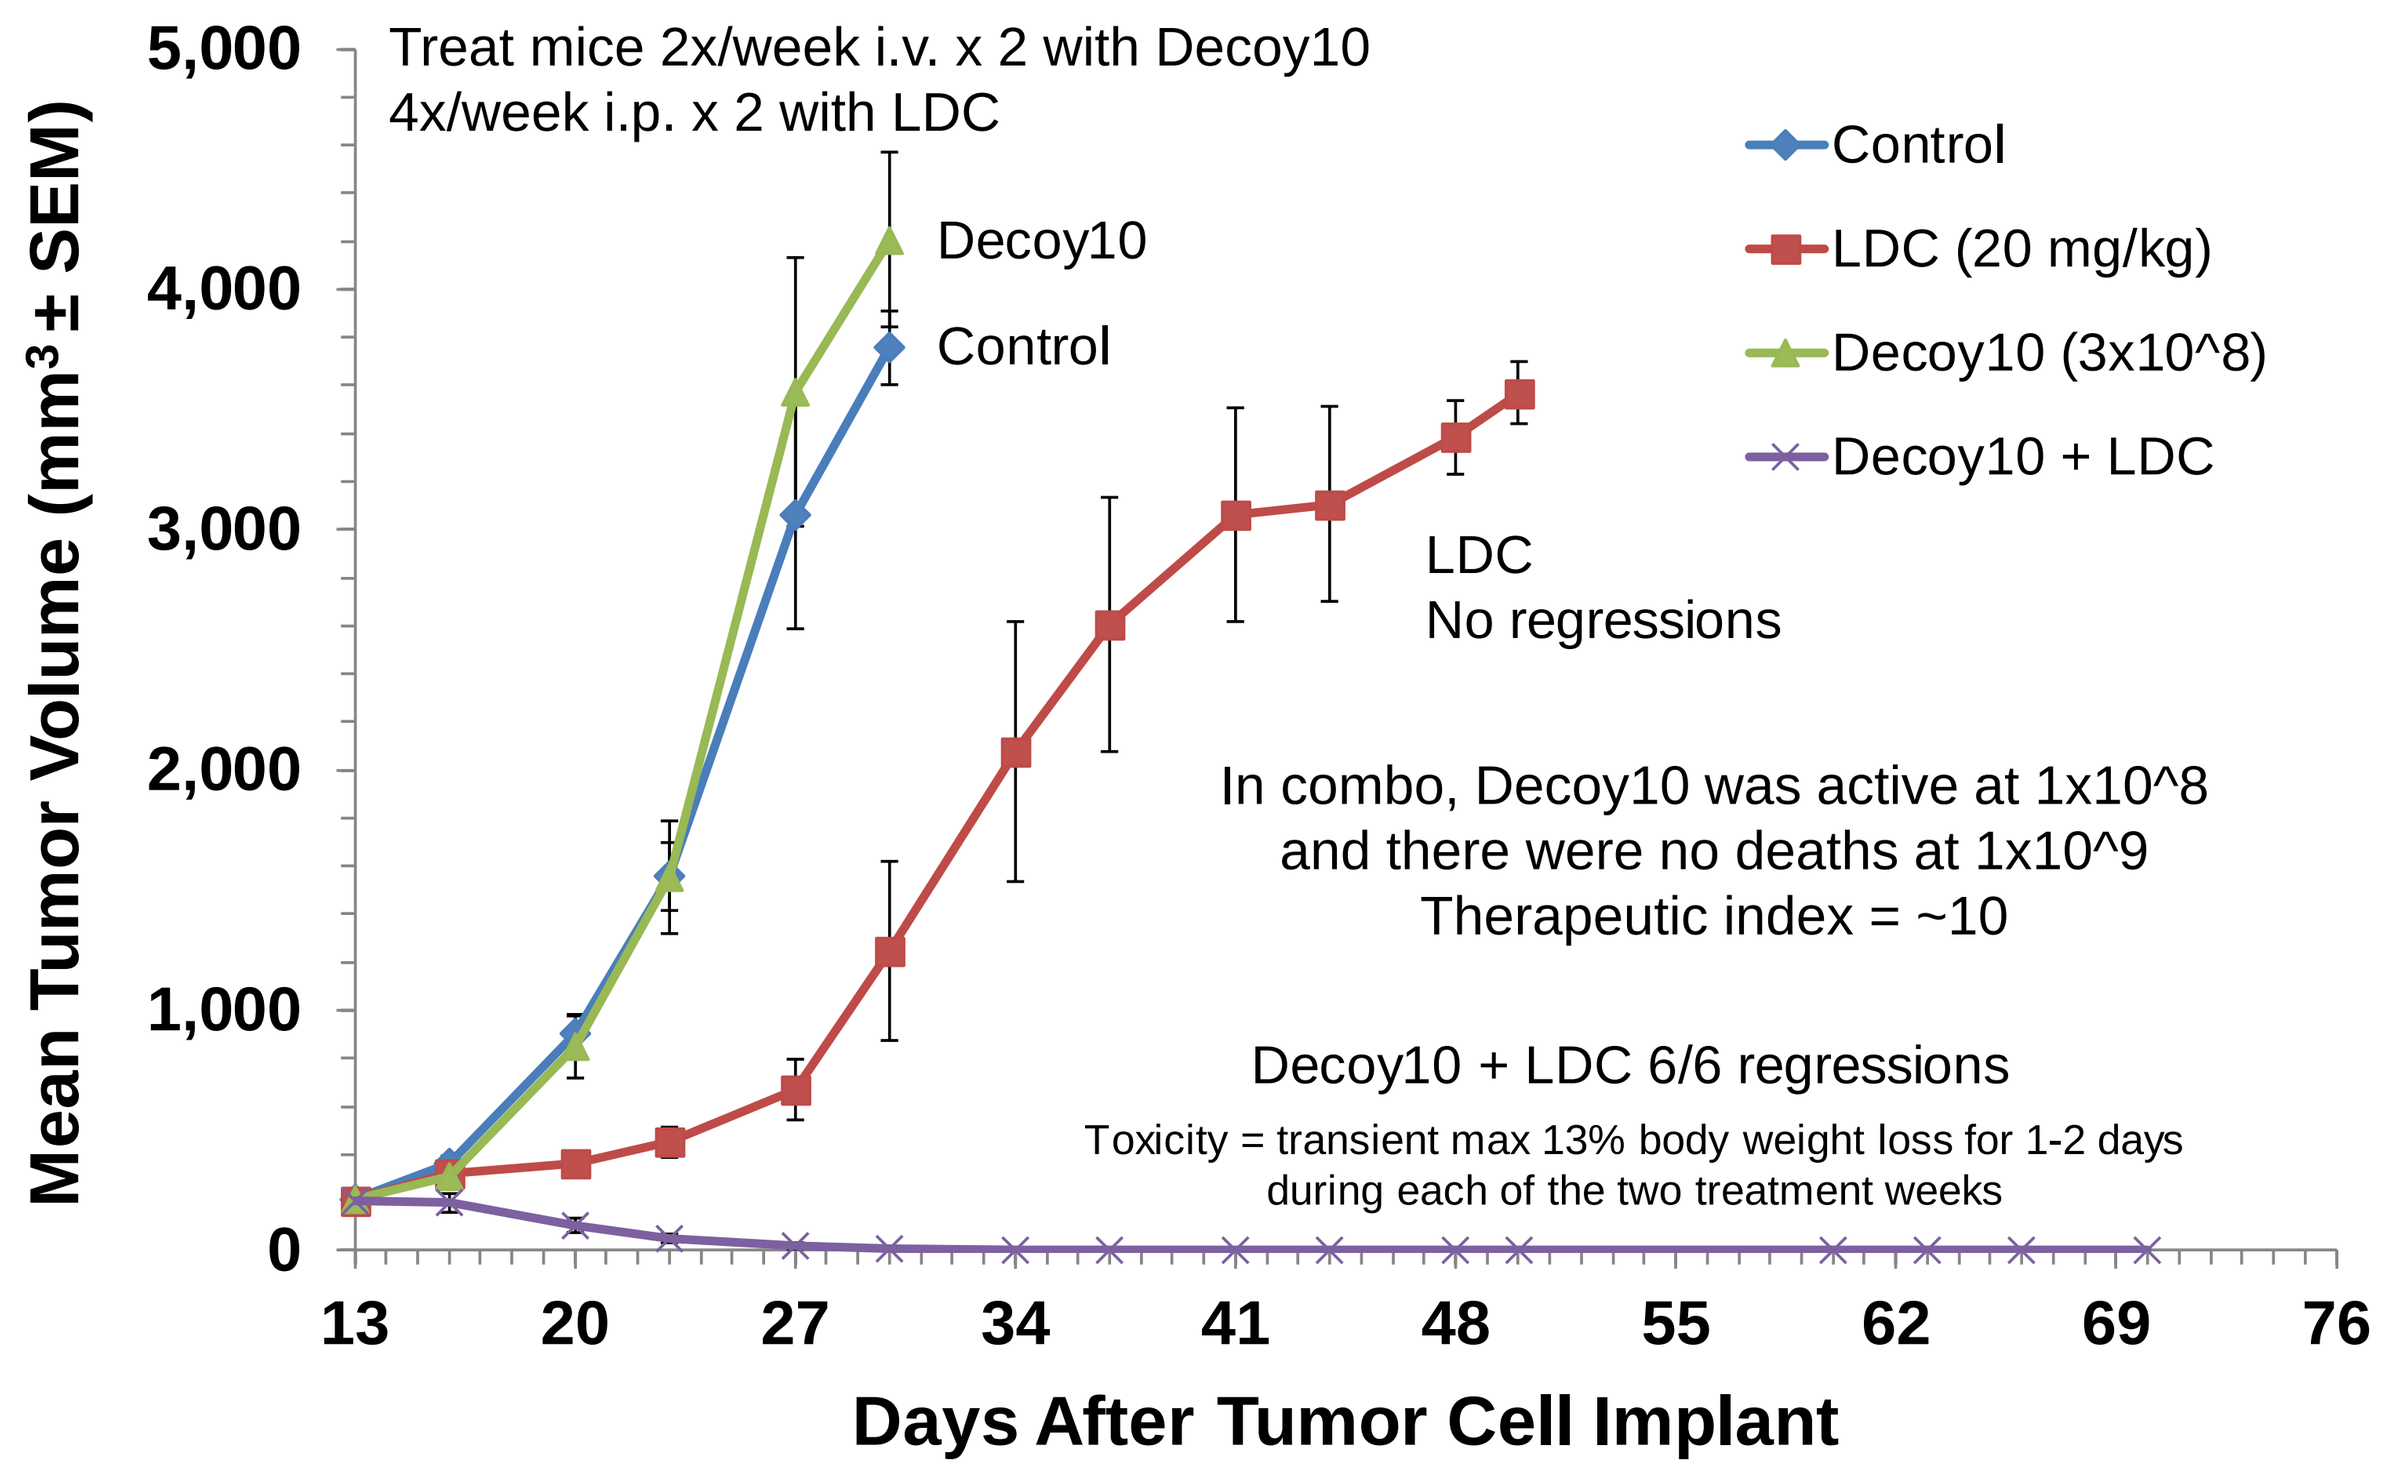
Mean Tumor Volume (mm³ ± SEM)
Treat mice 2x/week i.v. x 2 with Decoy10
4x/week i.p. x 2 with LDC
Control
LDC (20 mg/kg)
Decoy10 (3x10^8)
Decoy10 + LDC
Decoy10
Control
LDC
No regressions
In combo, Decoy10 was active at 1x10^8 and there were no deaths at 1x10^9
Therapeutic index = ~10
Decoy10 + LDC 6/6 regressions
Toxicity = transient max 13% body weight loss for 1-2 days during each of the two treatment weeks
Days After Tumor Cell Implant

Figure 4. Synergistic eradication of established NHL by Decoy technology is reproducible, durable and induces immunological memory

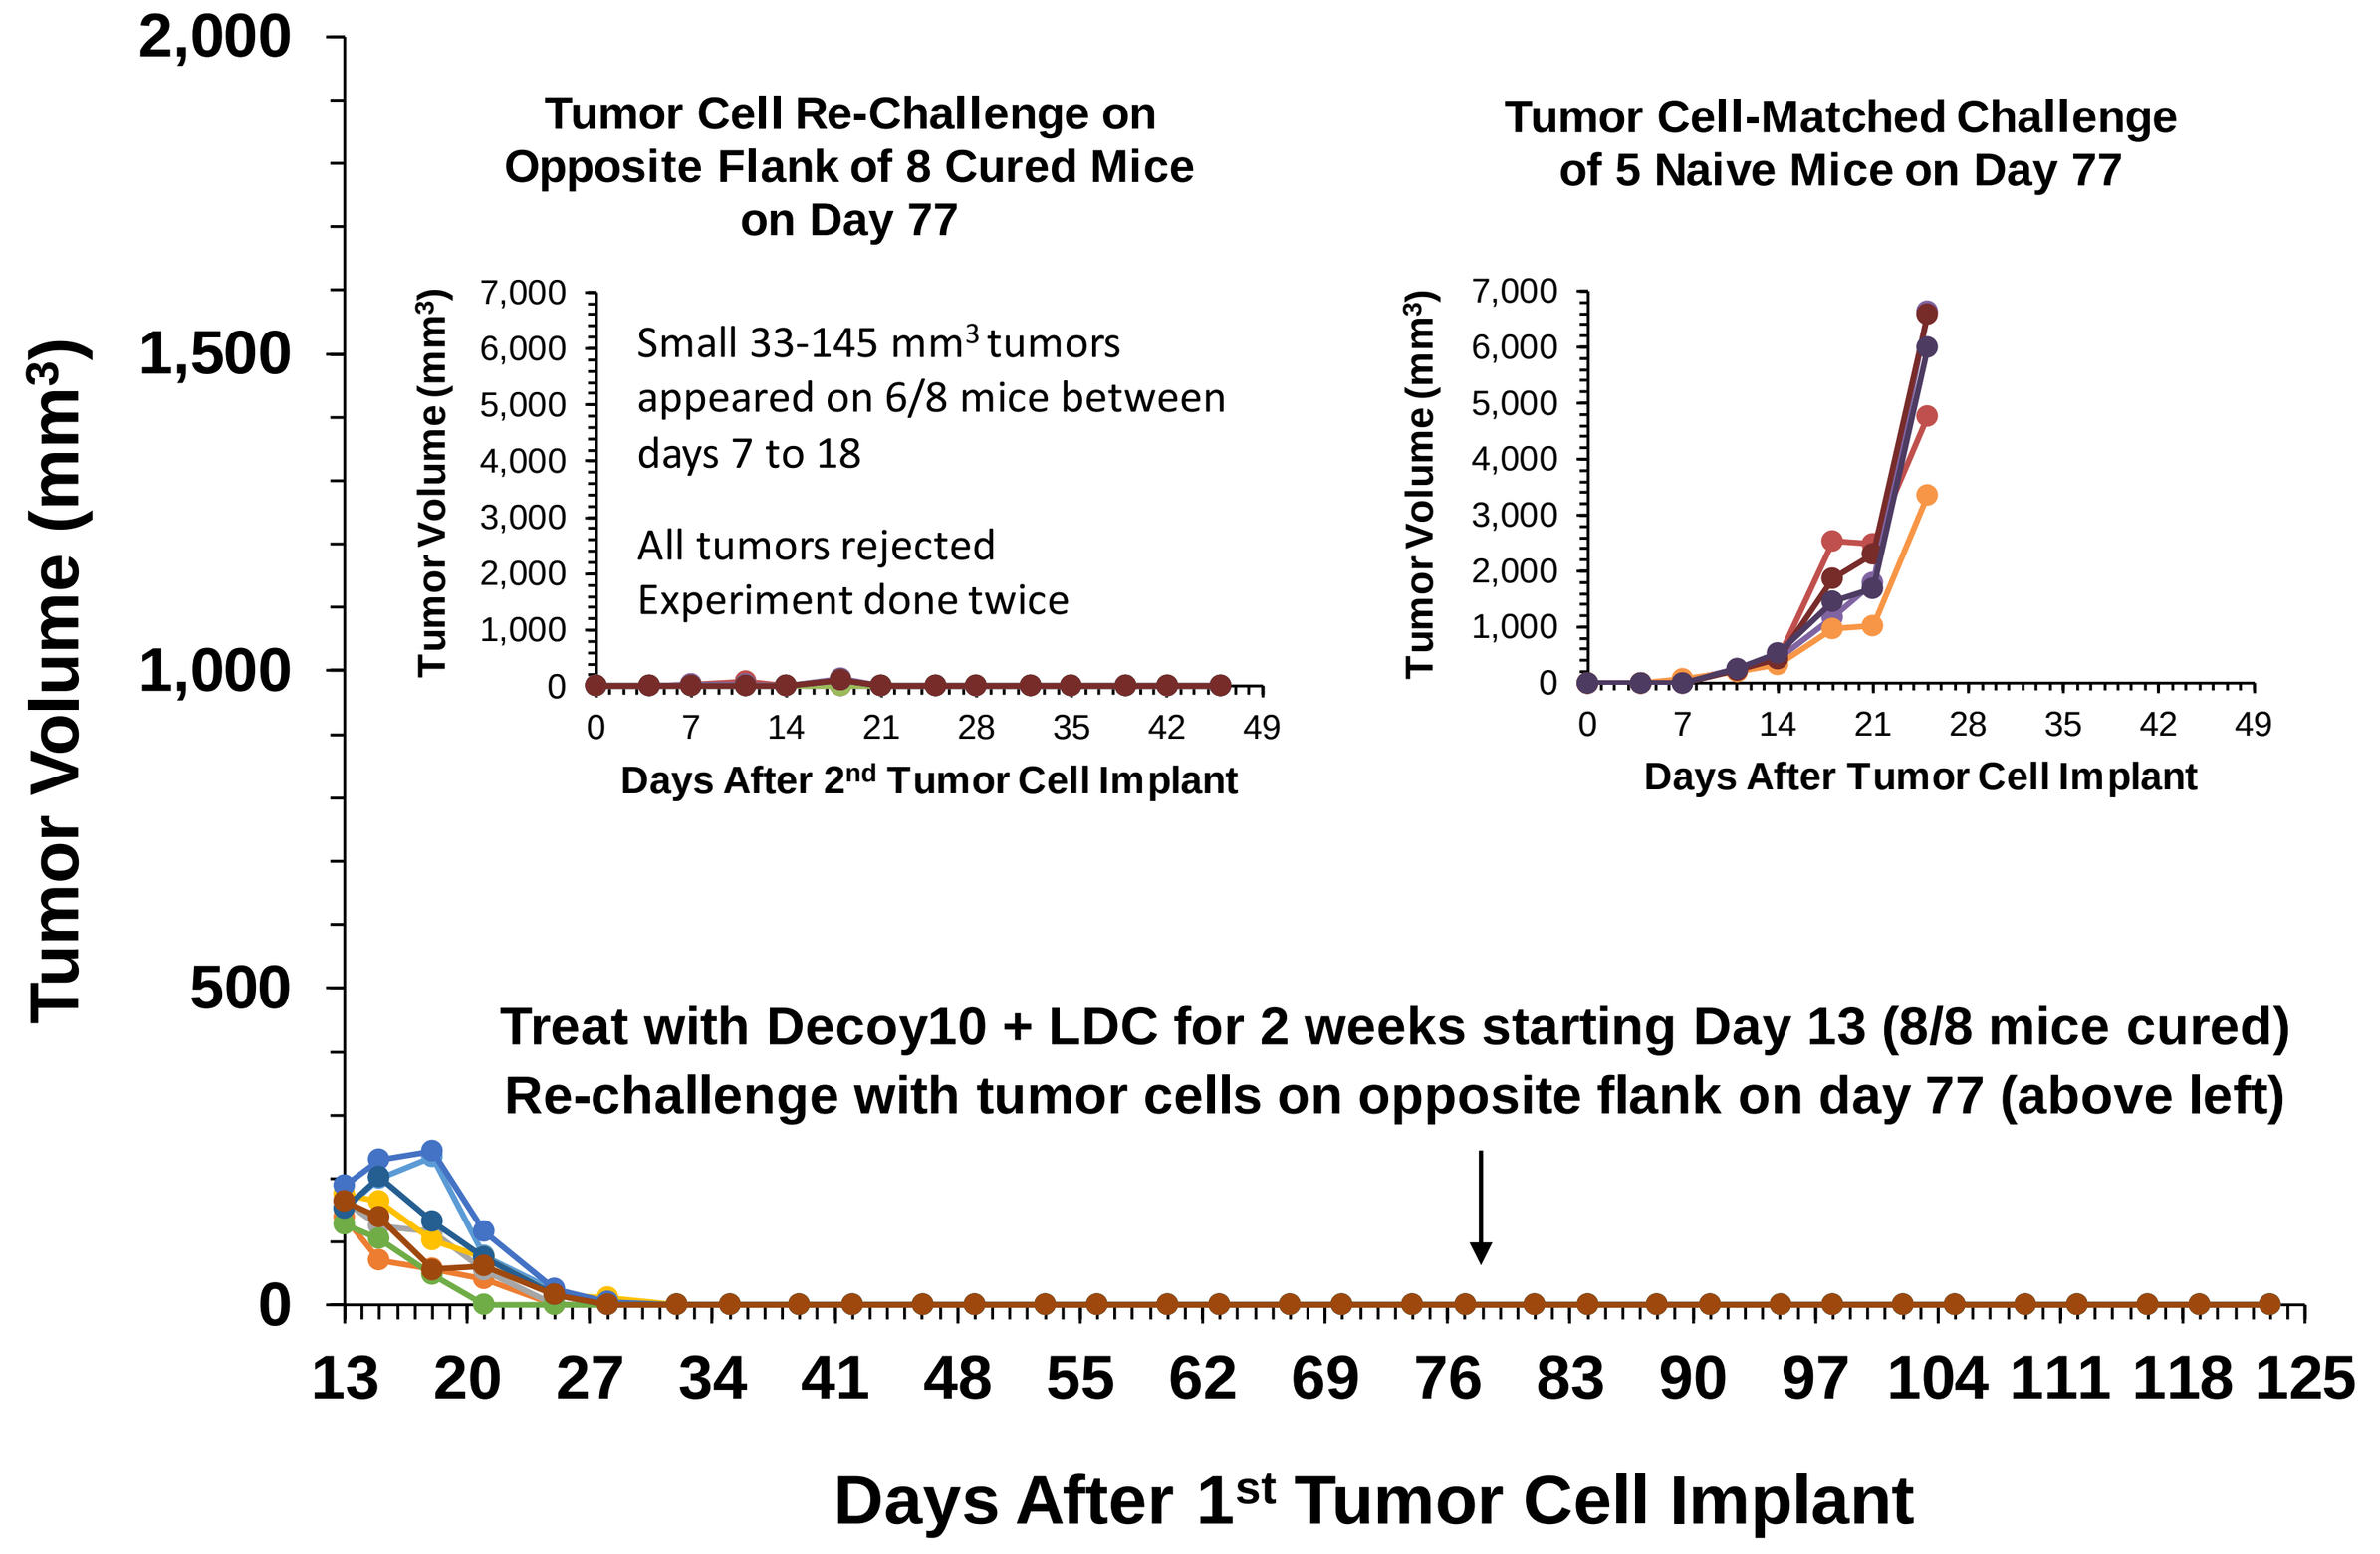
Tumor Volume (mm³)
Tumor Cell Re-Challenge on Opposite Flank of 8 Cured Mice on Day 77
Small 33-145 mm³ tumors appeared on 6/8 mice between days 7 to 18
All tumors rejected
Experiment done twice
Days After 2nd Tumor Cell Implant
Tumor Cell-Matched Challenge of 5 Naive Mice on Day 77
Days After Tumor Cell Implant
Treat with Decoy10 + LDC for 2 weeks starting Day 13 (8/8 mice cured)
Re-challenge with tumor cells on opposite flank on day 77 (above left)
Days After 1st Tumor Cell Implant

Results

Figure 5. Mouse A20 NHL tumors that regrow after sub-optimal Decoy treatment are sensitive to optimal re-treatment (very large tumors eradicated)

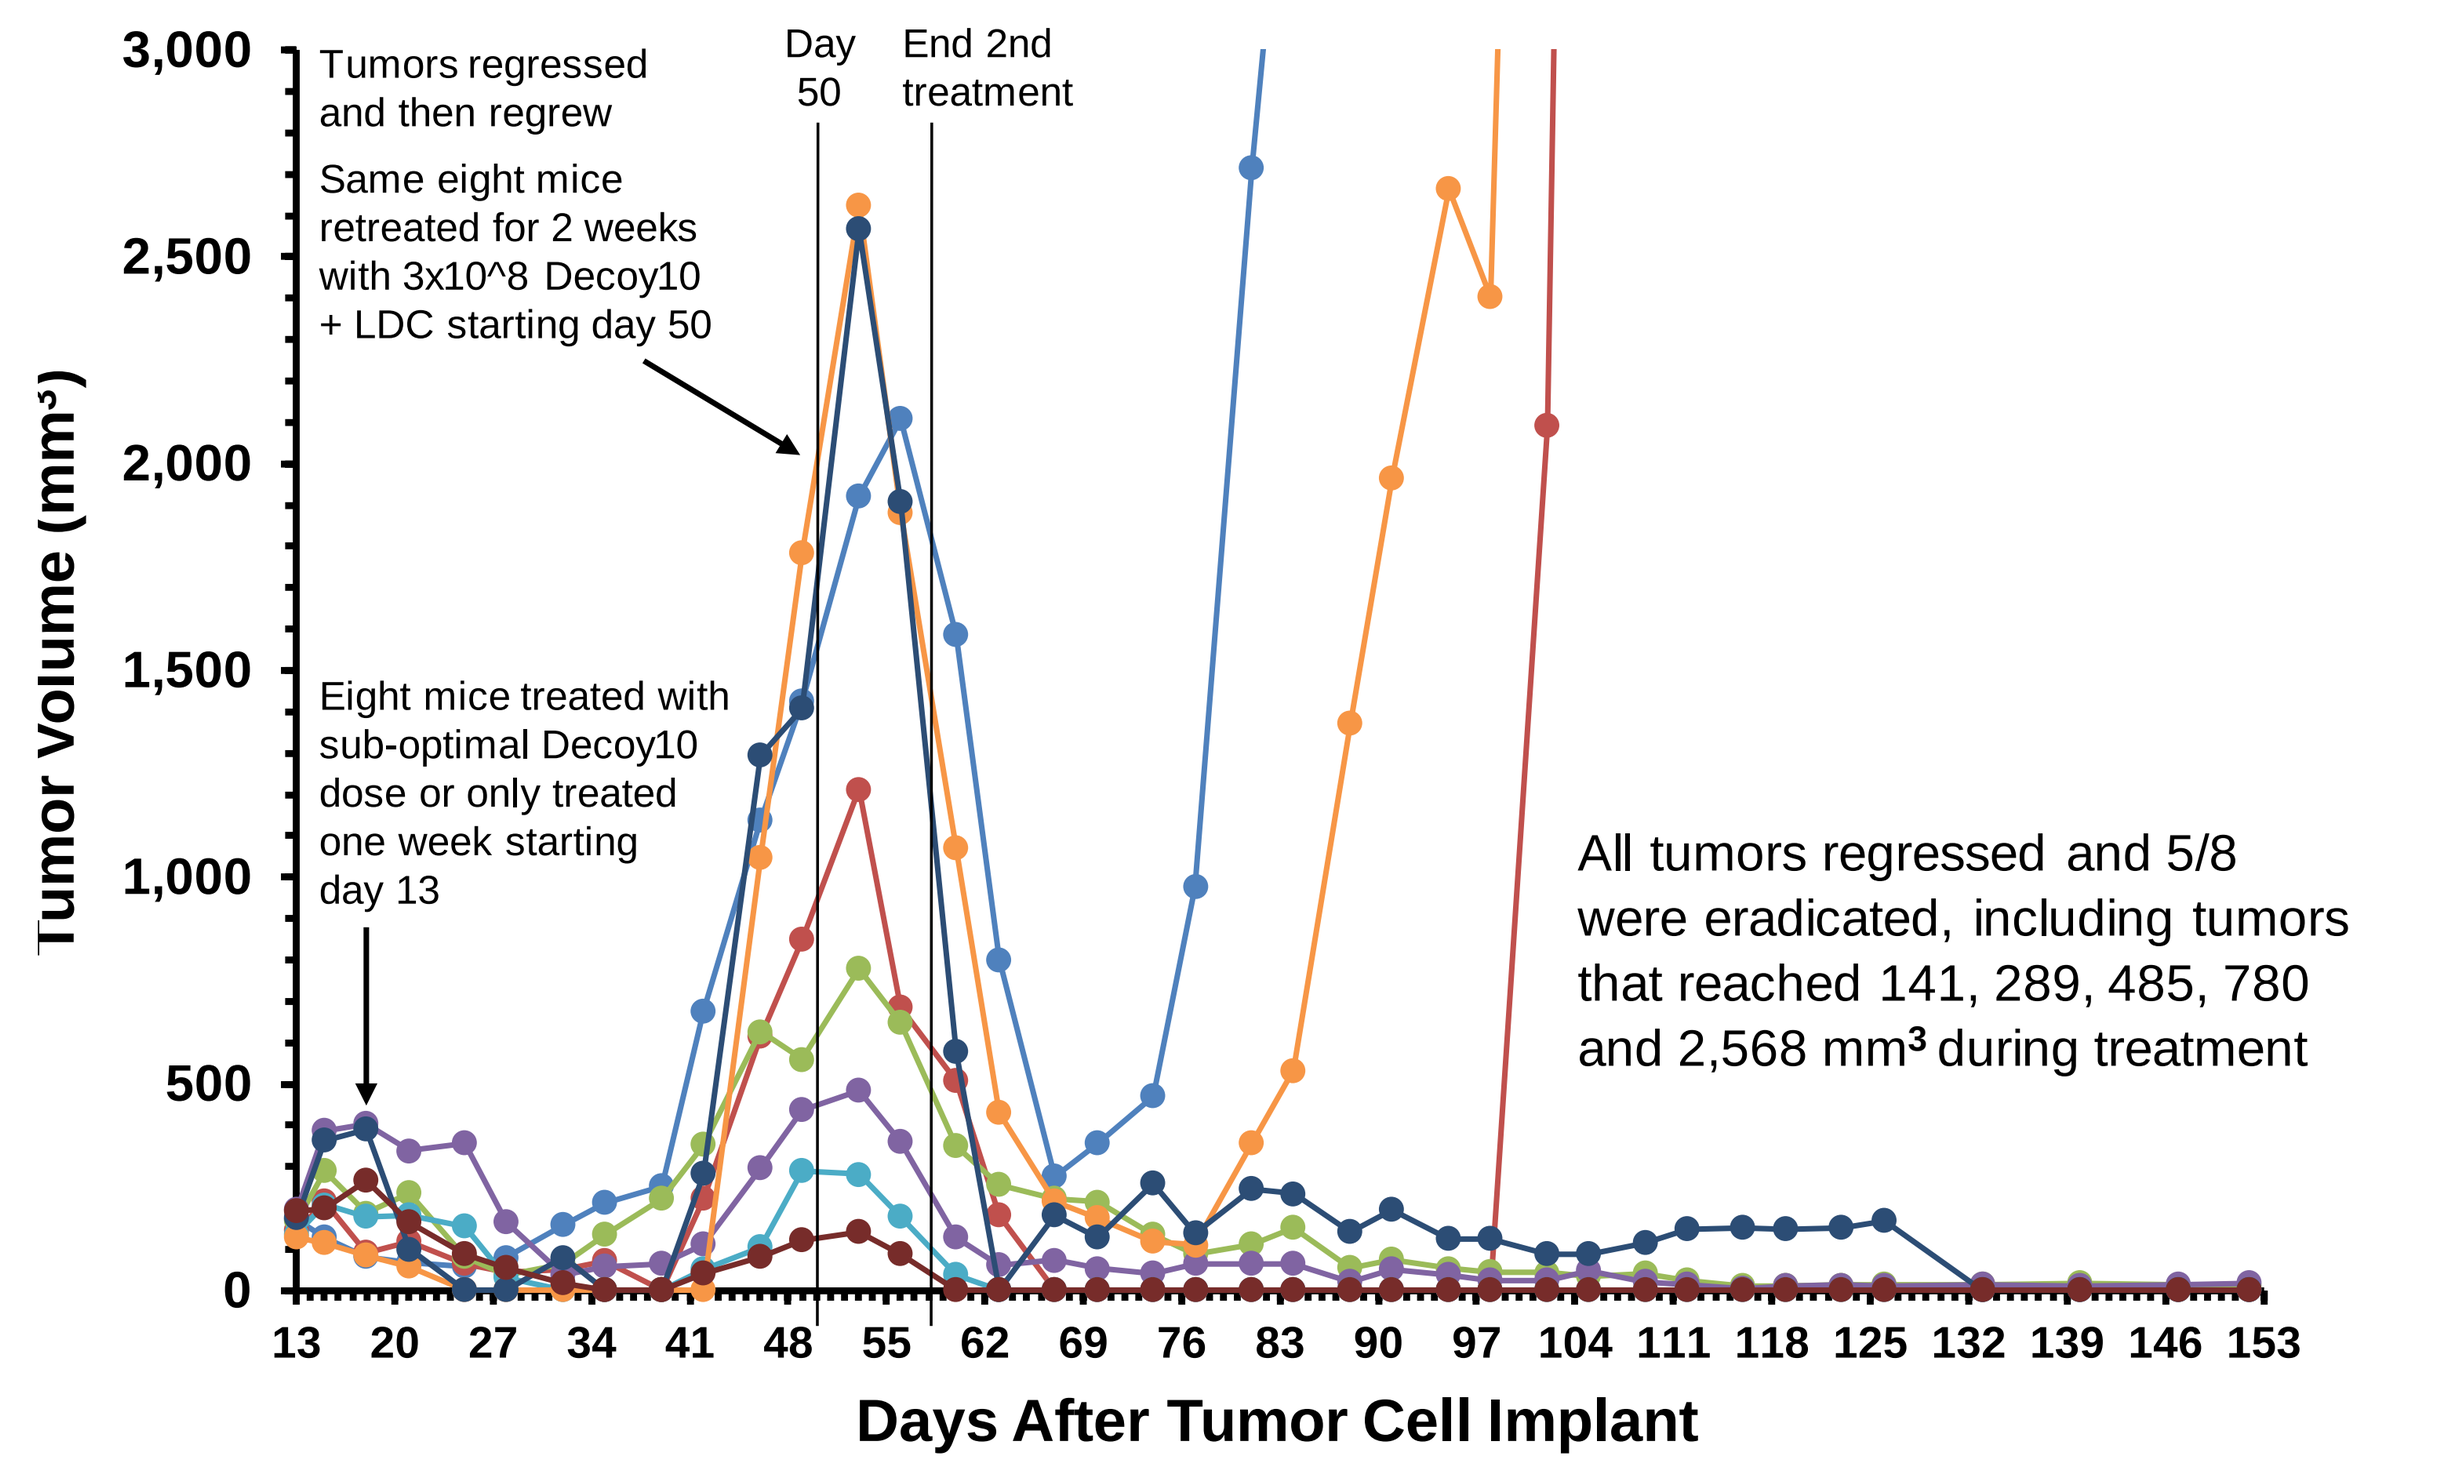
Tumor Volume (mm³)
Tumors regressed and then regrew
Same eight mice retreated for 2 weeks with 3x10^8 Decoy10 + LDC starting day 50
Day 50
End 2nd treatment
Eight mice treated with sub-optimal Decoy10 dose or only treated one week starting day 13
All tumors regressed and 5/8 were eradicated, including tumors that reached 141, 289, 485, 780 and 2,568 mm³ during treatment
Days After Tumor Cell Implant

Figure 6. NK cells, CD4+ and CD8+ T cells all participate in eradication of NHL by Decoy10 + LDC

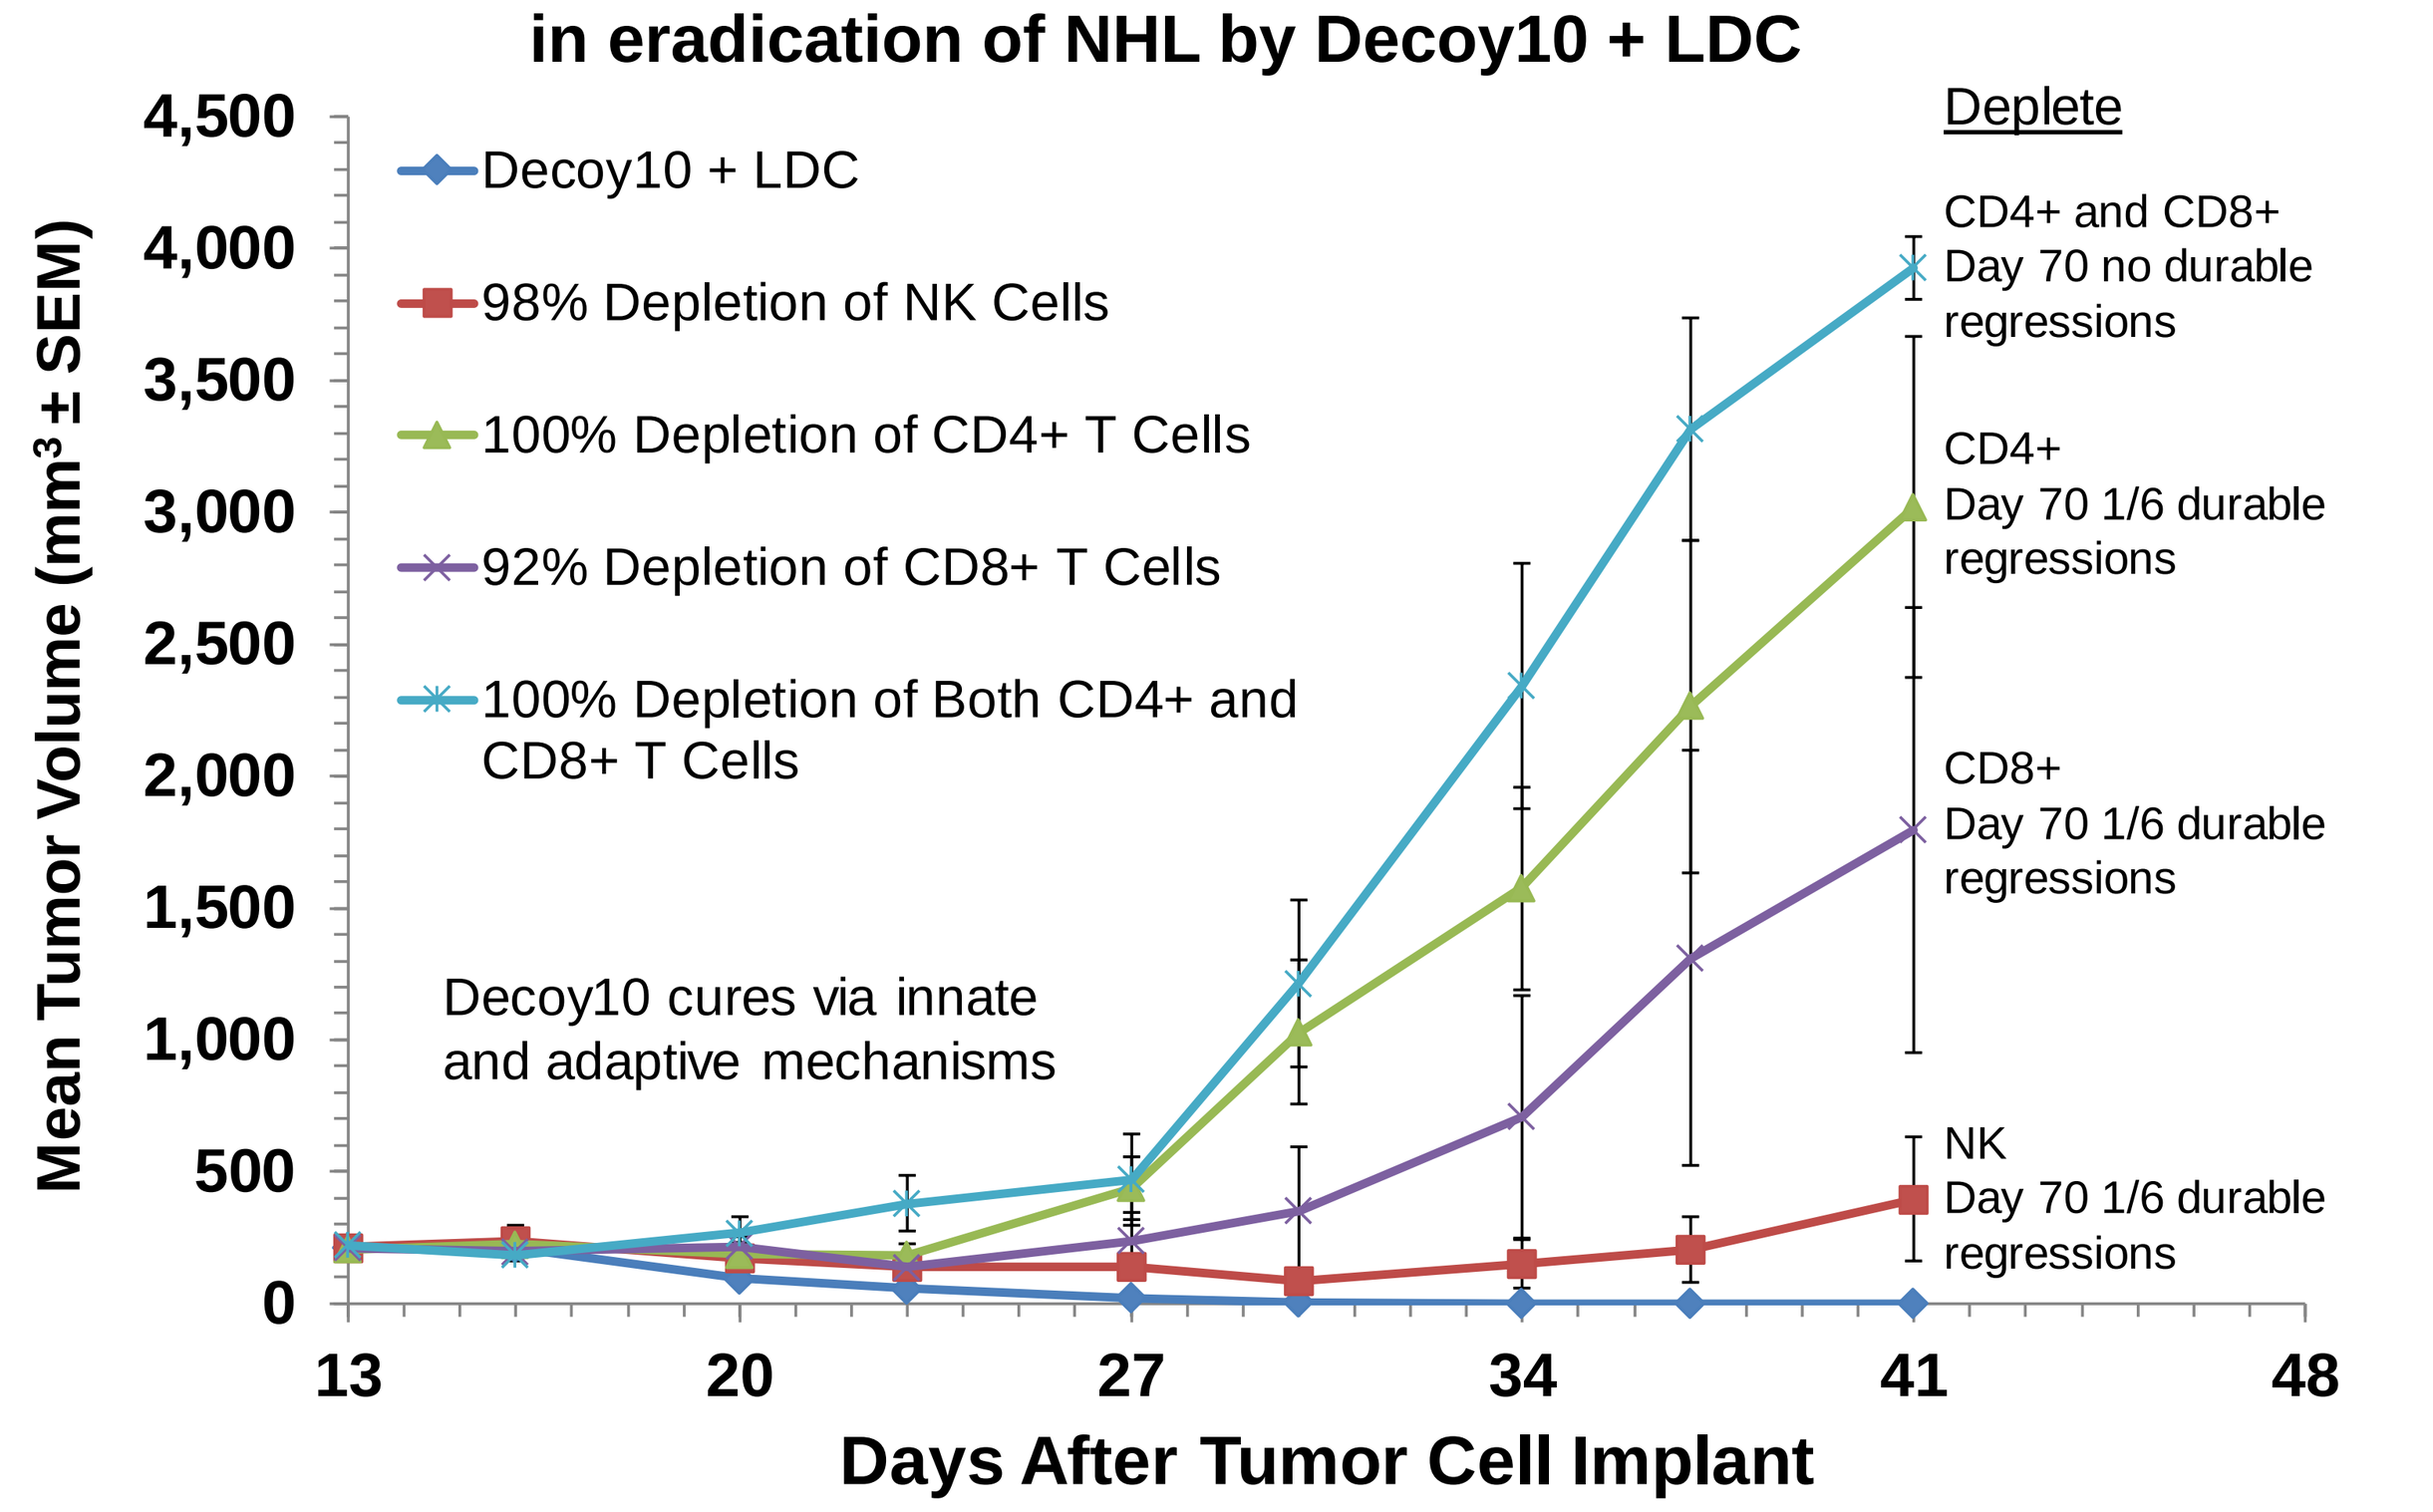
Mean Tumor Volume (mm³ ± SEM)
Decoy10 + LDC
98% Depletion of NK Cells
100% Depletion of CD4+ T Cells
92% Depletion of CD8+ T Cells
100% Depletion of Both CD4+ and CD8+ T Cells
Decoy10 cures via innate and adaptive mechanisms
Deplete
CD4+ and CD8+ Day 70 no durable regressions
CD4+ Day 70 1/6 durable regressions
CD8+ Day 70 1/6 durable regressions
NK Day 70 1/6 durable regressions
Days After Tumor Cell Implant

Figure 7. Decoy technology synergizes with rituximab to induce regression of s.c. human Ramos NHL in SCID mice

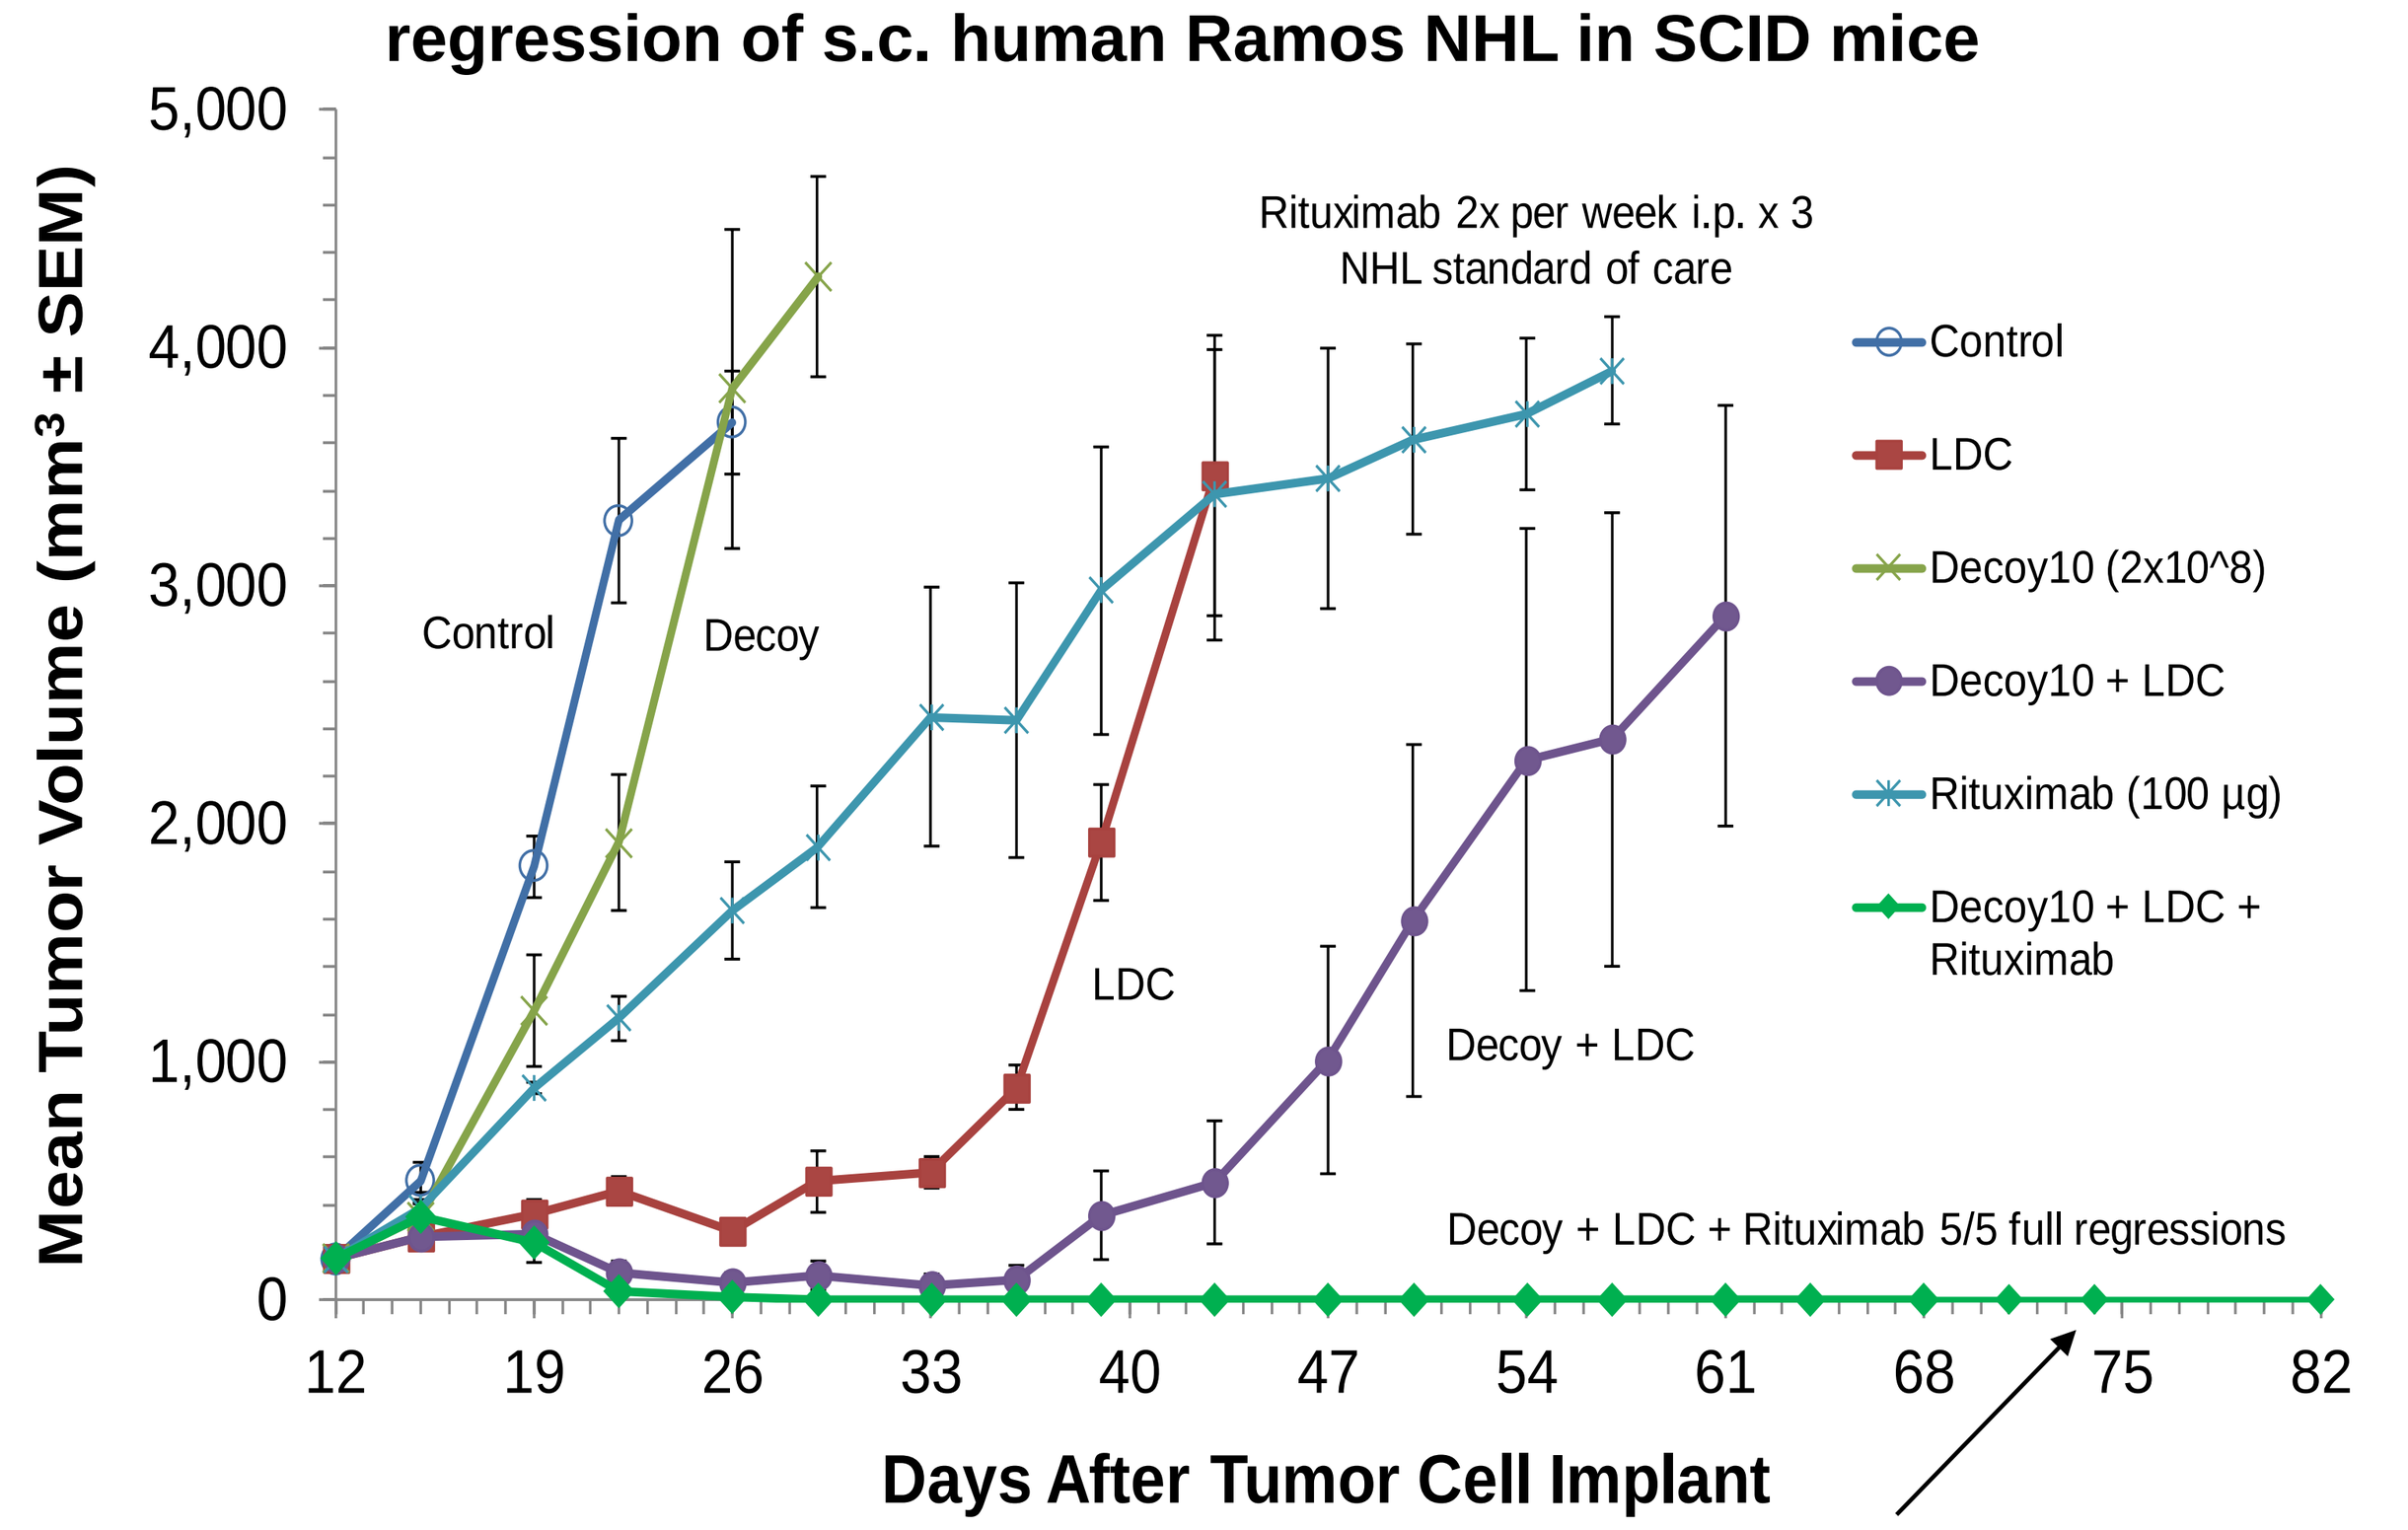
Mean Tumor Volume (mm³ ± SEM)
Rituximab 2x per week i.p. x 3
NHL standard of care
Control
LDC
Decoy10 (2x10^8)
Decoy10 + LDC
Rituximab (100 µg)
Decoy10 + LDC + Rituximab
Control
Decoy
LDC
Decoy + LDC
Decoy + LDC + Rituximab 5/5 full regressions
Days After Tumor Cell Implant

Re-challenged with fresh tumor cells on day 74 – 3/5 new tumors rejected

Results

Figure 8. Decoy10 synergizes with low-dose indomethacin to regress 4/6 established 186 mm³ s.c. mouse H22 hepatocellular carcinomas

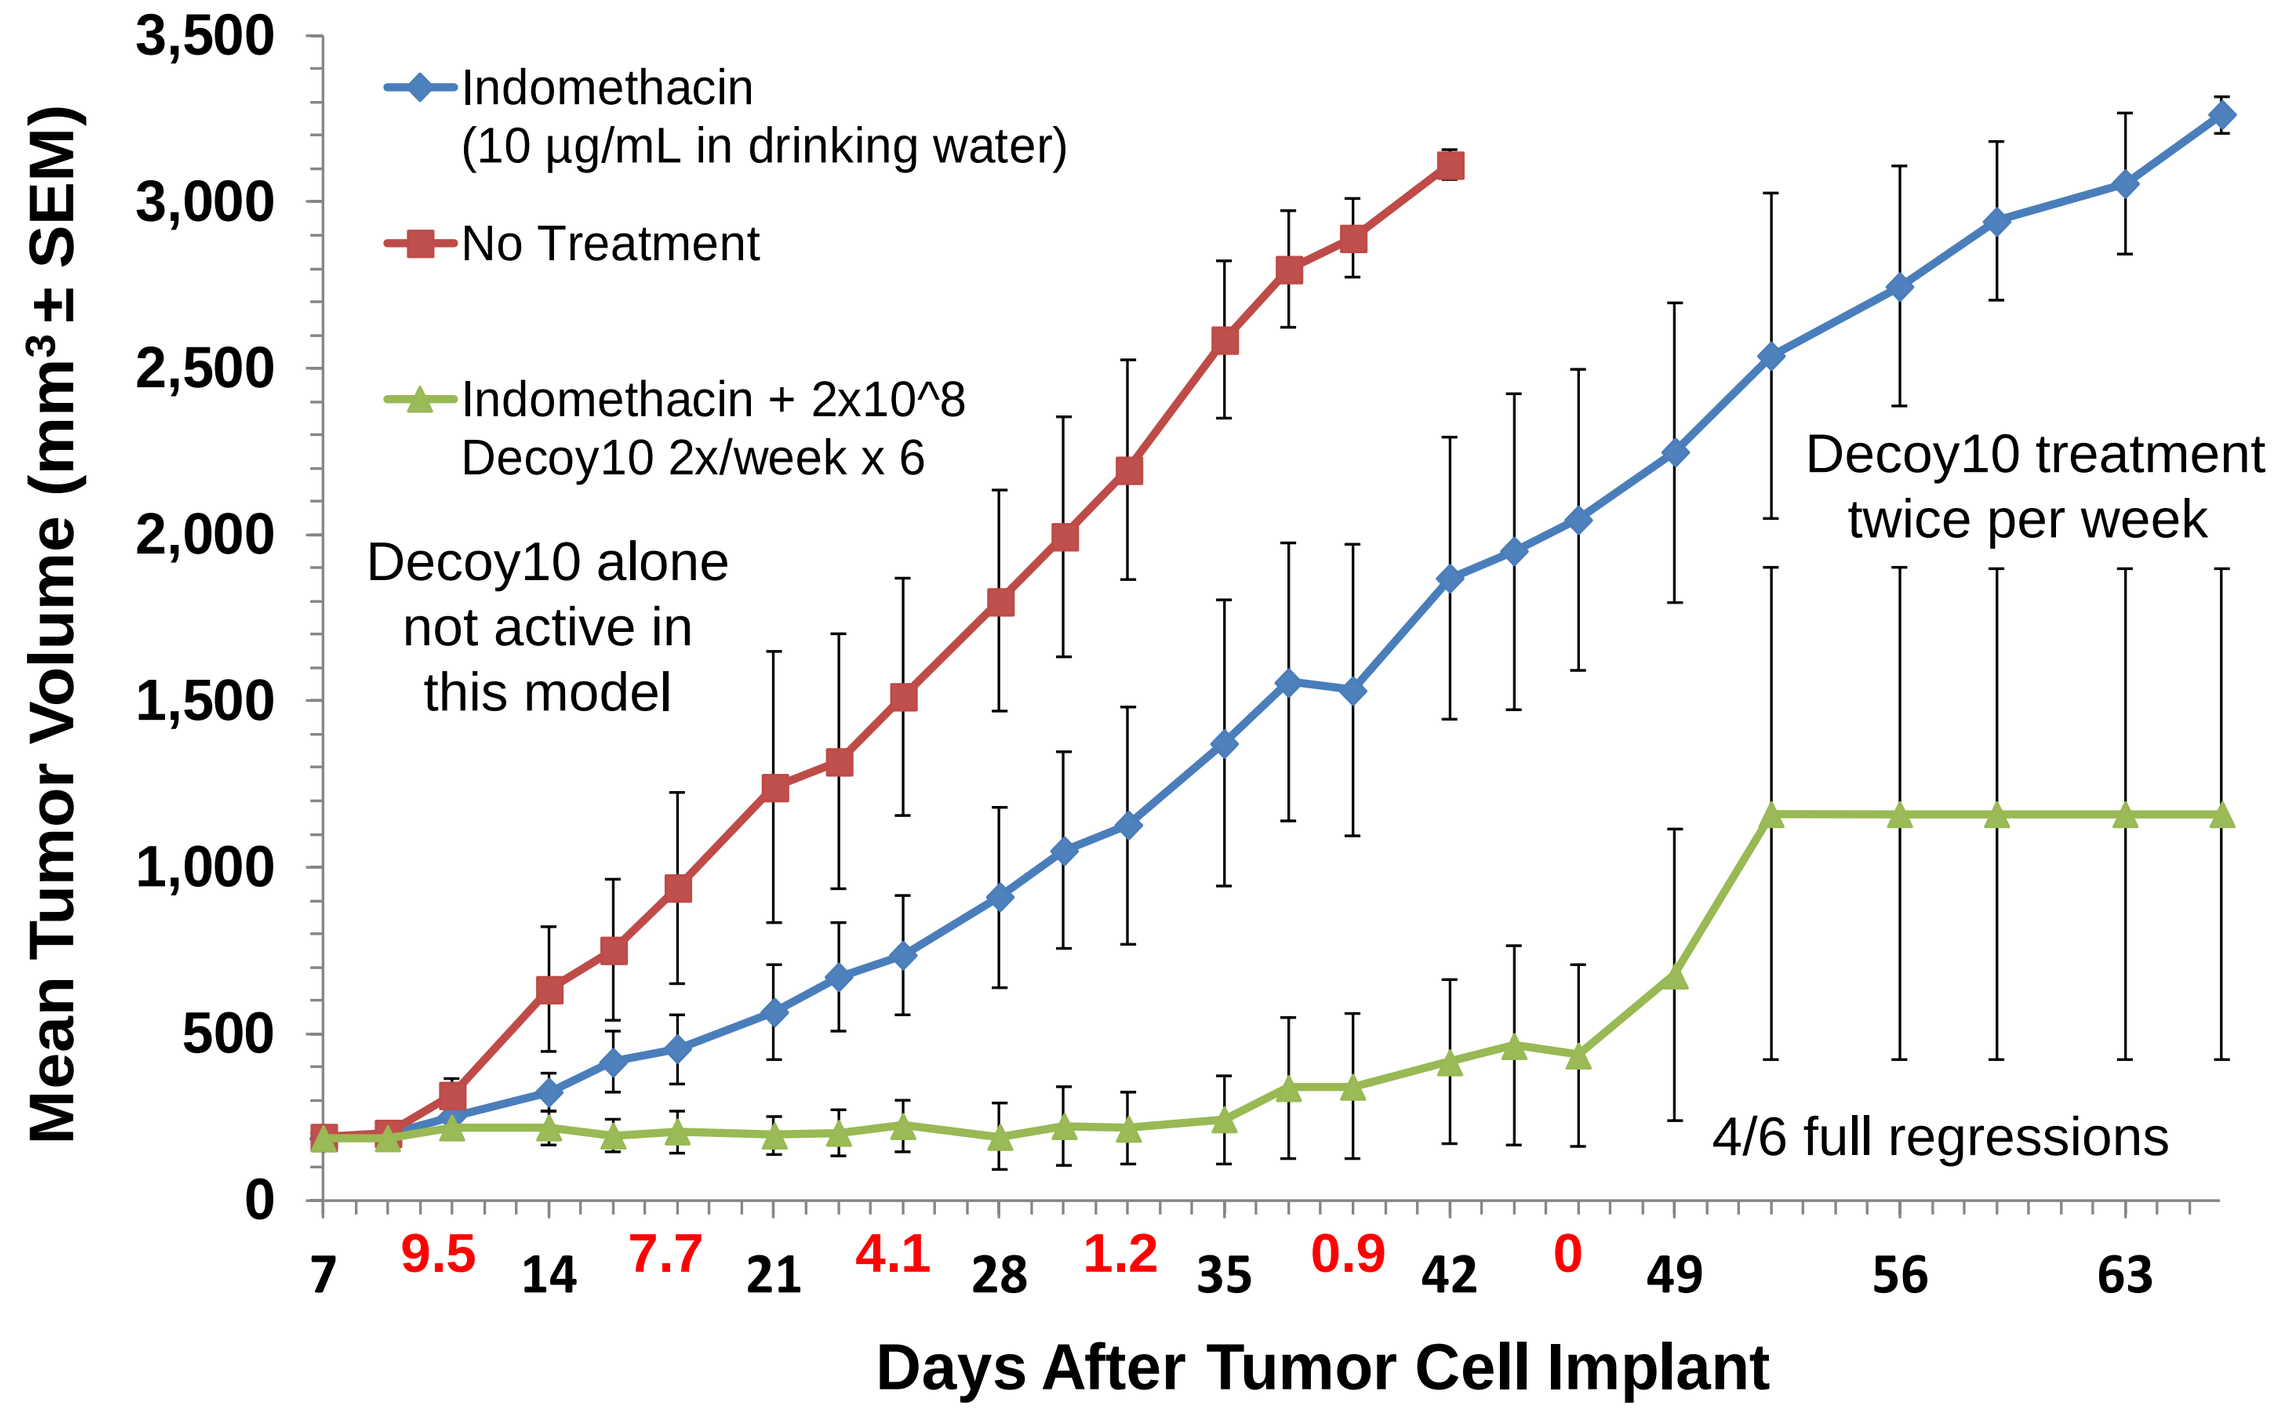
Mean Tumor Volume (mm³ ± SEM)
Indomethacin
(10 µg/mL in drinking water)
No Treatment
Indomethacin + 2x10^8
Decoy10 2x/week x 6
Decoy10 alone not active in this model
Decoy10 treatment twice per week
4/6 full regressions
9.5 7.7 4.1 1.2 0.9 0
Days After Tumor Cell Implant

Maximum % weight loss during each week of combo treatment in red above

Figure 9. Decoy10 synergizes with anti-PD-1 to regress 100% of established 186 mm³ s.c. mouse H22 hepatocellular carcinomas

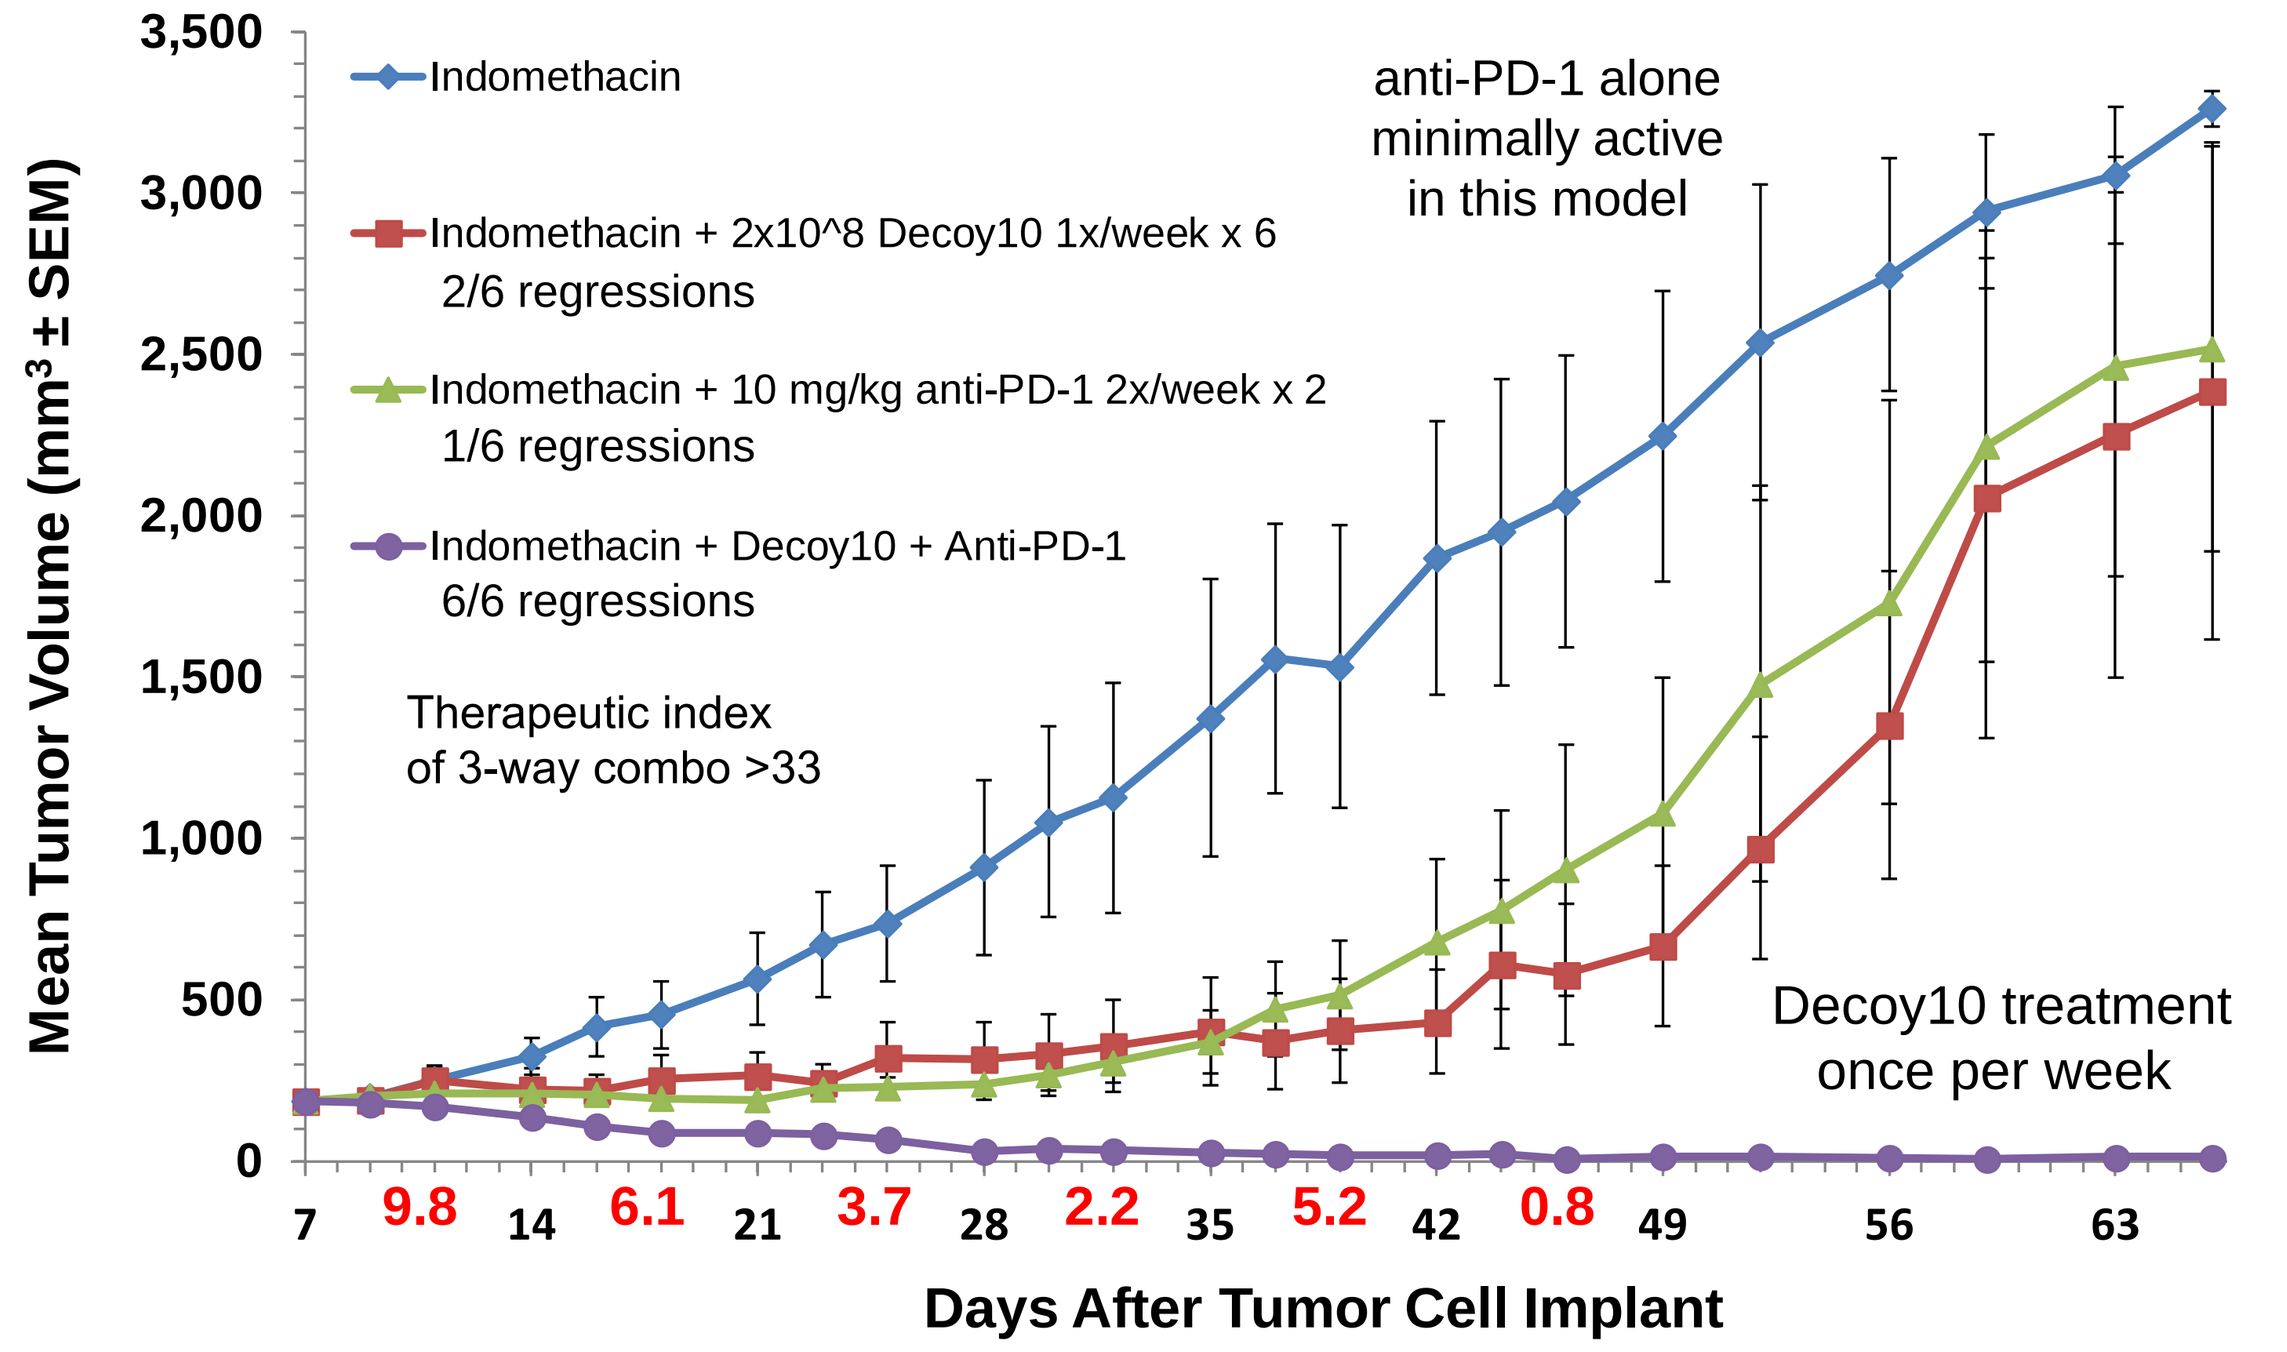
Mean Tumor Volume (mm³ ± SEM)
Indomethacin
Indomethacin + 2x10^8 Decoy10 1x/week x 6
2/6 regressions
Indomethacin + 10 mg/kg anti-PD-1 2x/week x 2
1/6 regressions
Indomethacin + Decoy10 + Anti-PD-1
6/6 regressions
anti-PD-1 alone minimally active in this model
Therapeutic index of 3-way combo >33
Decoy10 treatment once per week
9.8 6.1 3.7 2.2 5.2 0.8
Days After Tumor Cell Implant

Maximum % weight loss during each week of combo treatment in red above

Summary

We have produced a killed, multiple TLR agonist product from a single strain of non-pathogenic, Gram-negative bacteria with reduced *in vivo* i.v. toxicity, but without compromised immune system activation, as measured by induction of cytokine secretion by PBMCs (Decoy10 - Improved Coley's Toxins).

Decoy10 induces significant single agent and combination i.v. anti-tumor activity, including eradication of established tumors, with immunological memory, via synergy with each of five different approved agents (IL-2, low-dose chemotherapy, low-dose NSAID, rituximab and anti-PD-1 checkpoint therapy). The therapeutic index of Decoy bacteria in combination with LDC was ~10 and was >33 with anti-PD-1.

Decoy technology is active against colorectal, hepatocellular, pancreatic carcinoma and non-Hodgkin's lymphoma (so far) and eradicates tumors via activation of both innate and adaptive pathways.